Combining Oral OTC Analgesics: Analysis for an Intentional Drug Development Approach

P82

Robin Mathew[1], Sergey Motov[2], Sandy Tian[1], Jeffrey Gudin[3], James A McGuire[1], Daniel Budnitz[1], Alankar Gupta[1]

[1]Kenvue Brands LLC, Fort Washington, PA; [2]Maimonides Medical Center, Brooklyn, NY; [3]Englewood Hospital and Medical Center, Englewood, NJ

Introduction

According to a 2023-2024 national survey, acetaminophen and the nonsteroidal anti-inflammatory drugs (NSAIDs) aspirin, ibuprofen, and naproxen sodium are among the most commonly used medications in the United States.[1]

Current federal and professional society guidelines recommend non-opioid therapies as first-line treatment for many pain conditions and support multimodal analgesia, including the combined use of an NSAID and acetaminophen, to improve pain control while reducing opioid use.[2-6] However, combining multiple analgesics may increase regimen complexity and pill burden highlighting the need for rational development of analgesic combinations.

Selecting a non-opioid fixed-dose combination (FDC) that optimizes pharmacologic complementarity may address this need by providing the advantages of multimodal analgesia while also reducing pill burden, increasing convenience, and potentially reducing patient medication administration errors.

Objective

To assess **pharmacologic complementarity** and identify **potential combinations** of oral OTC analgesics (acetaminophen, aspirin, ibuprofen, and naproxen sodium) for optimal multimodal pain management as an FDC:

- We reviewed published pharmacokinetic (PK) parameters
 - Time to maximum plasma concentration (T_{max})
 - Terminal elimination half-life ($t_{½}$)
- We describe reported mechanisms of action (MOA)

Methods

A systematic review of publications indexed on PubMed/MEDLINE was conducted for each oral OTC analgesic in April 2026 using the search terms:

- *((acetaminophen) OR (paracetamol)) AND (pharmacokinetics); aspirin AND pharmacokinetics; ibuprofen AND pharmacokinetics; and ("naproxen sodium"[tiab:~0]) AND (pharmacokinetics)*
- An English language filter was applied to all searches.

Primary references cited by review articles were evaluated for inclusion. Inclusion criteria included:

- Human studies with ≥5 participants
- Reported T_{max} and/or $t_{½}$
- Single oral dose in tablet or capsule form
- Taken in fasting state

Publications were excluded if they included:

- Only a pediatric population
- Prescription doses
- Non-tablet, multidose, or multidrug administrations
- Non-oral formulations
- Non-healthy volunteers (eg, renal or hepatic disease, diabetes mellitus, etc.)
- Did not report design, sample size, or fed/fasted state

All publications identified were exported to EndNote 21 for further review.

Data extracted were T_{max} and $t_{½}$, reported as a mean, median, geometric mean, or least squares mean along with standard deviation, standard error, or geometric coefficient of variation.

MOA were identified from manufacturer monographs/product reference information.

Results

Flow Diagram of Literature Search Strategy

PubMed search (English language filter applied)

1251 publications total:
- 576 acetaminophen
- 351 aspirin
- 294 ibuprofen
- 75 naproxen sodium

Removal of non-relevant and secondary references

102 publications for full-text review:
- 46 acetaminophen
- 18 aspirin
- 33 ibuprofen
- 5 naproxen sodium

Removal due to lack of PK data or study design elements

75 publications included:
- 35 acetaminophen
- 14 aspirin
- 26 ibuprofen
- 4 naproxen sodium*

**Fewer naproxen sodium studies reflect its limited formulations/doses*

Butterfly Plot of Publications Reporting T_{max} and $t_{½}$ for OTC Acetaminophen and NSAIDs

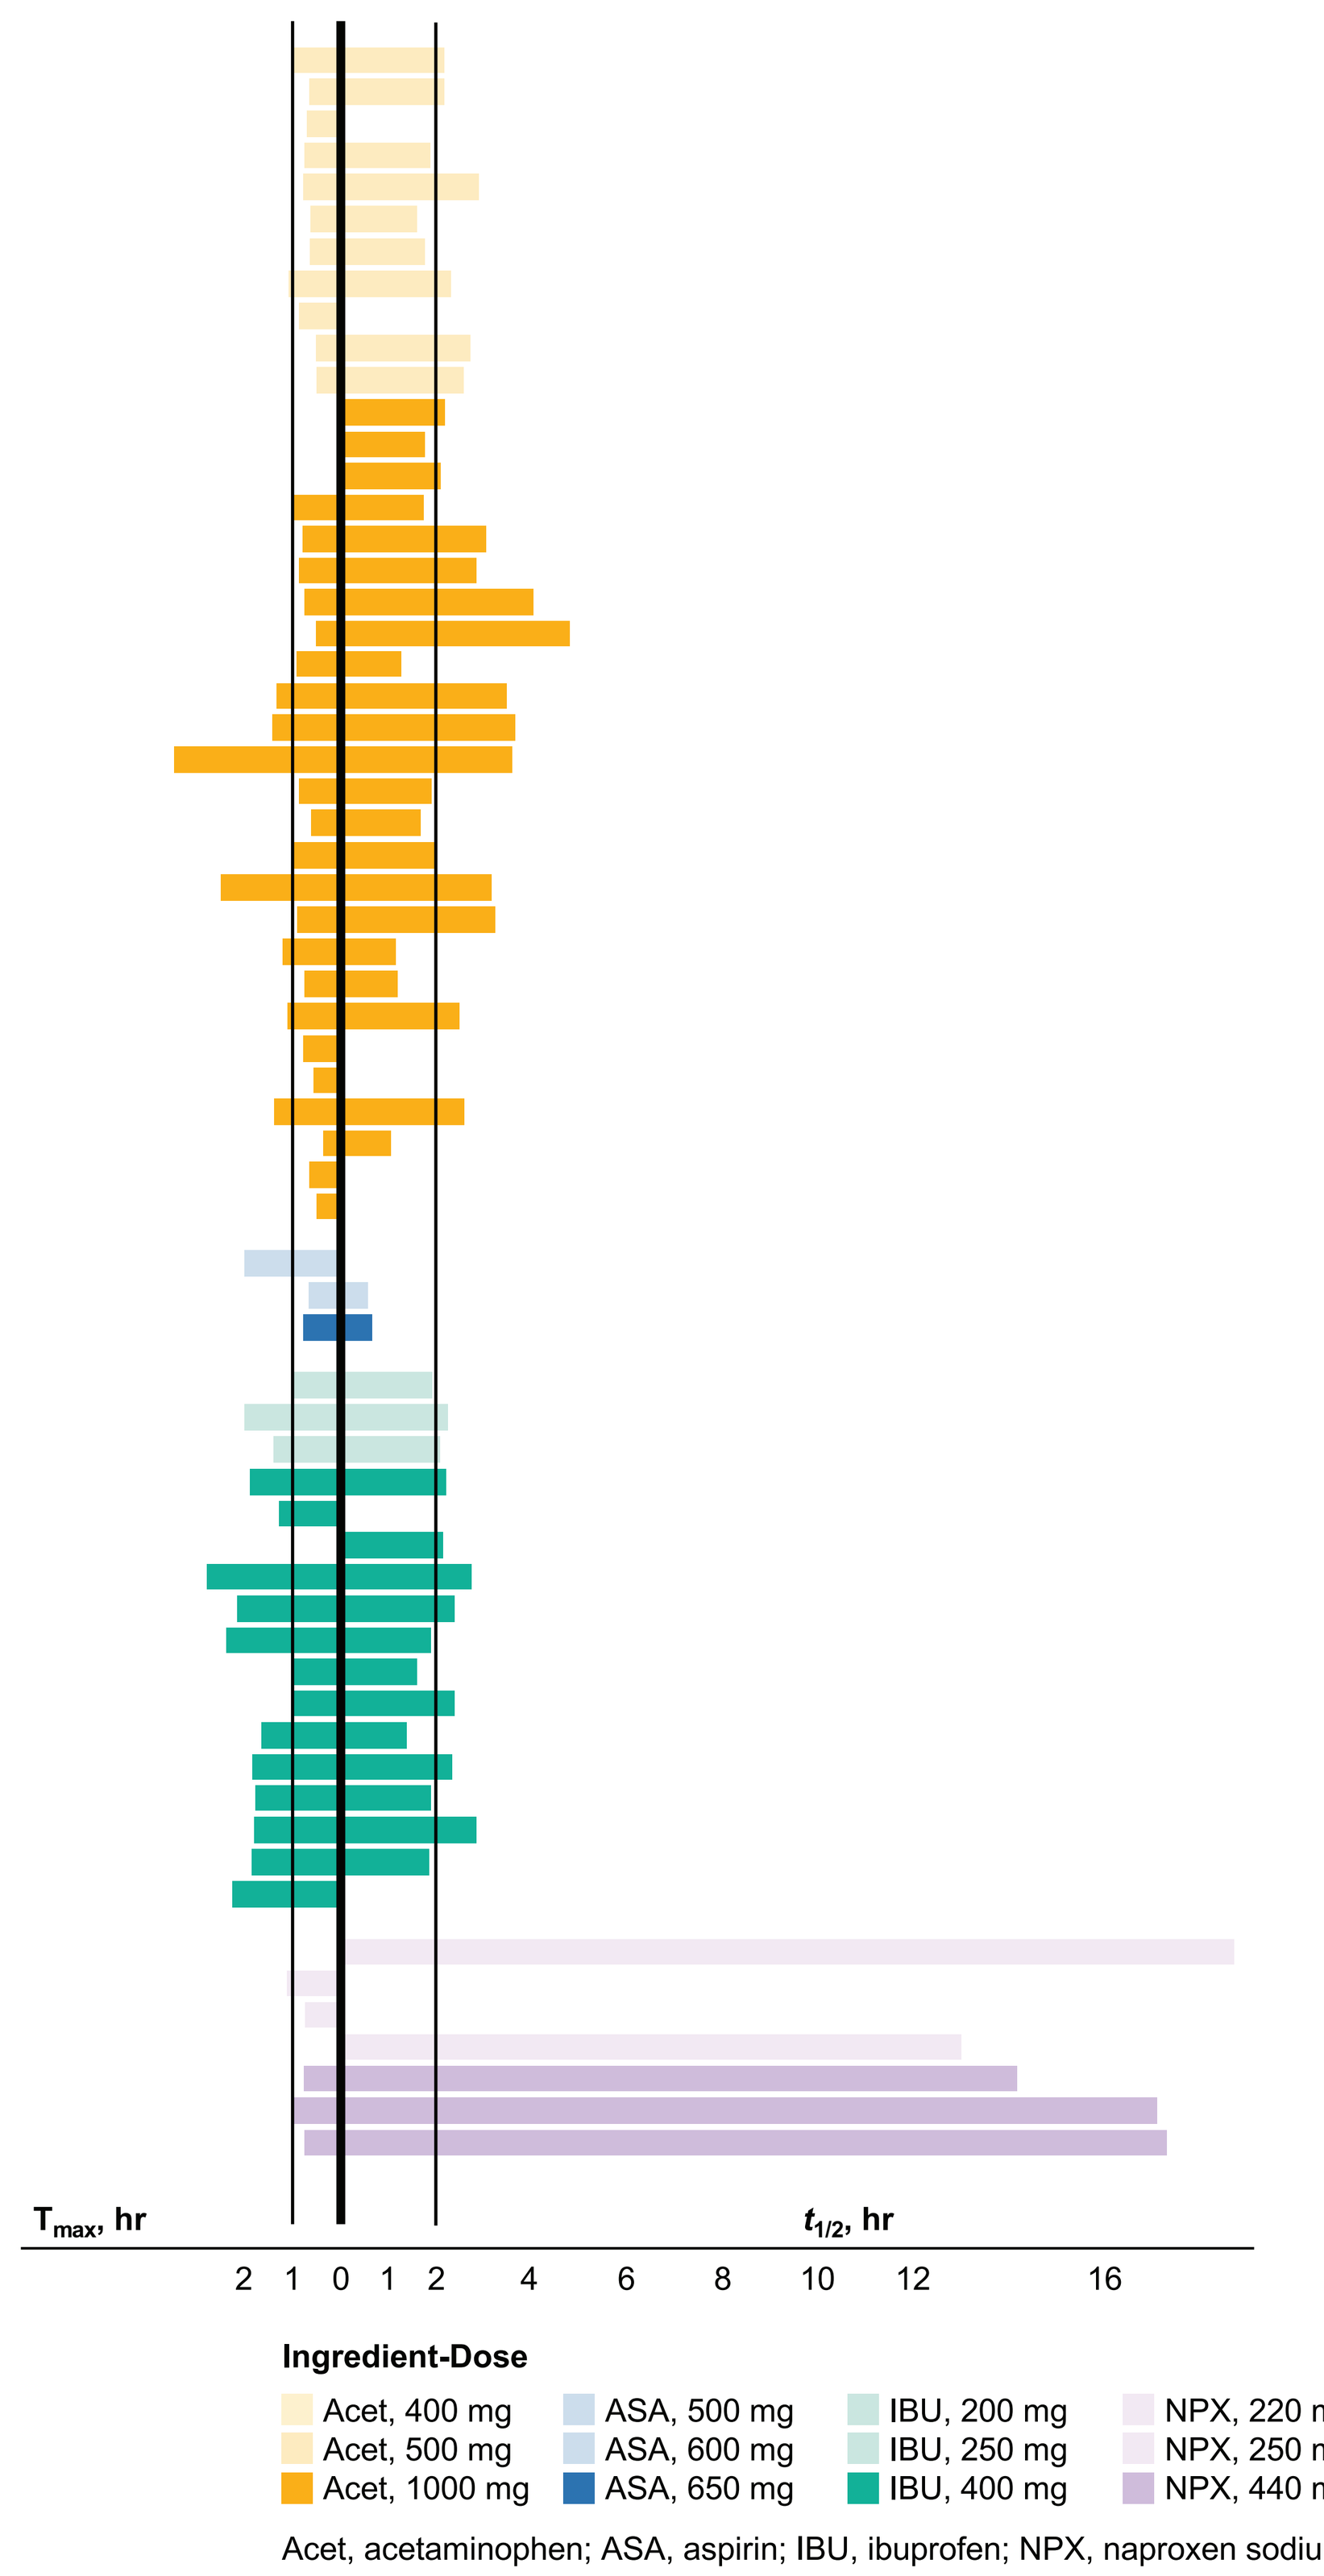
T_{max}, hr
$t_{1/2}$, hr
2 1 0 1 2 4 6 8 10 12 16
Ingredient-Dose
Acet, 400 mg
Acet, 500 mg
Acet, 1000 mg
ASA, 500 mg
ASA, 600 mg
ASA, 650 mg
IBU, 200 mg
IBU, 250 mg
IBU, 400 mg
NPX, 220 mg
NPX, 250 mg
NPX, 440 mg

Agent	Dose	Year	Study	Design	N
Acet	400 mg	1997	Muir	Cross-over	12
Acet	500 mg	1991	Rumble	Cross-over	8
Acet	500 mg	1991	Seideman	Cross-over	10
Acet	500 mg	1995	Iqbal	Cross-over	10
Acet	500 mg	2001	Yin	Single arm	12
Acet	500 mg	2002	Schaiquevich-WO saliva	Cross-over	5
Acet	500 mg	2002	Schaiquevich-With saliva	Cross-over	5
Acet	500 mg	2017	Hammami	Cross-over	50
Acet	500 mg	2018	Idkaidek	Parallel	10
Acet	500 mg	2020	Tarabar	Cross-over	44
Acet	500 mg	2024	Harder	Cross-over	14
Acet	1000 mg	1976	Briant-older	Parallel	28
Acet	1000 mg	1976	Briant-younger age (20-40 y)	Parallel	28
Acet	1000 mg	1979	Andreasen	Parallel	12
Acet	1000 mg	1984	Dietz	Parallel	8
Acet	1000 mg	1986	Rayburn	Single arm	6
Acet	1000 mg	1993	Girre	Parallel	12
Acet	1000 mg	1997	Seaber	Cross-over	15
Acet	1000 mg	2000	Strangier	Cross-over	12
Acet	1000 mg	2001	Loniewski	Cross-over	10
Acet	1000 mg	2002	Gugilla-during follicular phase	Cross-over	12
Acet	1000 mg	2002	Gugilla-during luteal phase	Cross-over	12
Acet	1000 mg	2002	Gugilla-during ovulation	Cross-over	12
Acet	1000 mg	2002	Kolawole-at 1:00pm	Cross-over	7
Acet	1000 mg	2002	Kolawole-at 7:30am	Cross-over	7
Acet	1000 mg	2002	Kolawole-at 9:00pm	Cross-over	7
Acet	1000 mg	2003	Gandia	Cross-over	18
Acet	1000 mg	2004	Babalola	Single arm	8
Acet	1000 mg	2004	Obodozie	Cross-over	5
Acet	1000 mg	2006	Issa	Cross-over	10
Acet	1000 mg	2006	Tankanltelr	Parallel	16
Acet	1000 mg	2007	Renner	Cross-over	24
Acet	1000 mg	2007	Shinoda	Parallel	5
Acet	1000 mg	2010	Chiew	Cross-over	9
Acet	1000 mg	2013	Issa	Cross-over	10
Acet	1000 mg	2018	Miles	Cross-over	30
Acet	1000 mg	2019	Jacob	Cross-over	6
ASA	500 mg	2020	Schick	Cross-over	30
ASA	600 mg	1997	Muir	Cross-over	12
ASA	650 mg	1999	Sagar	Cross-over	10
IBU	200 mg	1974	Kaiser	Cross-over	20
IBU	200 mg	2020	Tarabar-study 1	Cross-over	34
IBU	250 mg	2020	Tarabar-study 2	Cross-over	44
IBU	400 mg	1983	Juhl	Cross-over	29
IBU	400 mg	1983	Lockwood	Cross-over	15
IBU	400 mg	1984	Albert	Cross-over	32
IBU	400 mg	1999	Bapuji-at 10:00am	Cross-over	8
IBU	400 mg	1999	Bapuji-at 10:00pm	Cross-over	8
IBU	400 mg	2004	Van Dyke	Parallel	32
IBU	400 mg	2006	Hynninen-R(-) enantiomer	Cross-over	12
IBU	400 mg	2006	Hynninen-S(+) enantiomer	Cross-over	12
IBU	400 mg	2008	Zheng-R(-) enantiomer	Cross-over	20
IBU	400 mg	2008	Zheng-S(+) enantiomer	Cross-over	20
IBU	400 mg	2008	Zheng-total/racemate	Cross-over	20
IBU	400 mg	2014	Clementi	Cross-over	12
IBU	400 mg	2017	Hammami	Cross-over	30
IBU	400 mg	2018	Miles	Cross-over	30
NPX	220 mg	2000	Toothaker	Cross-over; no variability measure reported	30
NPX	220 mg	2000	Toothaker-female	Cross-over; no variability measure reported	15
NPX	220 mg	2000	Toothaker-male	Cross-over; no variability measure reported	15
NPX	250 mg	1974	Runkel	Parallel	7
NPX	440 mg	2016	Laurora-fasted	Cross-over; fasted	28
NPX	440 mg	2019	Sugar-Bayer Aleve	Cross-over	18
NPX	440 mg	2019	Sugar-Reckitt test product	Cross-over	18

Acet, acetaminophen; ASA, aspirin; IBU, ibuprofen; NPX, naproxen sodium

General Patient Demographics and Characteristics

Age range	18–69 years
Body weight range	36–97 kg
Mean body mass index range	22.3–26.6 kg/m^2

Sample sizes of publications ranged from 5 to 50 subjects.

T_{max} and $t_{½}$ Summary for OTC Acetaminophen and NSAIDs

Substantial differences in $t_{½}$ between the ingredients was identified
- The **shortest** $t_{½}$ values were reported for **aspirin**: 0.58–0.66 hours*
- The **longest** $t_{½}$ values were reported for **naproxen sodium**: 13.0–18.7 hours
- $t_{½}$ values ranged from 1.1–4.8 hours for **acetaminophen** and 1.4–3.4 hours for **ibuprofen**

Reported T_{max} values showed more modest differences.
- 0.38–2.5 hours for **acetaminophen**
- 0.68–2.0 hours for **aspirin**
- 0.74–1.1 hours for **naproxen sodium**
- 1.0–2.8 hours for **ibuprofen**

**Reflects $t_{½}$ of the parental compound; salicylate, the active metabolite may persist longer (2–4.5 hours) at analgesic doses*

Reported MOA of Acetaminophen and NSAIDs[7-15]

Ingredient	MOA	Primary compartment
Acetaminophen	~18 potential mechanisms: multiple central mechanisms (serotonergic, endocannabinoid, TRPV1-related) with limited peripheral COX inhibition	Central (brain and CNS)
Aspirin	Non-selective COX-1/COX-2 inhibitor with slight preference for COX-1	Peripheral
Ibuprofen	Non-selective COX-1/COX-2 inhibitor with slight preference for COX-1	Peripheral
Naproxen sodium	Non-selective COX-1/COX-2 inhibitor	Peripheral

CNS, central nervous system; COX, cyclooxygenase

Conclusions

- Naproxen sodium and acetaminophen exhibit complementary PK profiles; the shorter half-life of acetaminophen potentially complements the longer half-life of naproxen sodium, supporting the potential for additive analgesia through sequential rather than overlapping plasma concentrations
- While many FDCs pair agents with similar T_{max} and $t_{½}$ values, a complementary PK approach may instead help mitigate peaks and troughs in pain control while maintaining multimodal analgesic activity.
- The complementary properties of acetaminophen and naproxen sodium may mimic key advantages of alternating analgesic regimens, providing both rapid analgesic onset and sustained duration of pain relief within a single treatment approach.
- Together, these pharmacologic characteristics support the potential for a simplified FDC regimen that may reduce pill burden, improve convenience, and decrease the risk of medication administration errors.
- Overall, the findings suggest a PK rationale for combining acetaminophen and naproxen sodium as a multimodal FDC capable of delivering both fast and durable pain relief while providing the adherence and usability advantages of a single product.

Limitations

- Included publications were identified using a relatively small number of search terms and thus may be limited by selection biases and publication bias.
- The exclusive use of PubMed potentially excluded some studies.
- There was a smaller number of publications for aspirin and naproxen sodium which may reflect limited formulations and doses, particularly for naproxen sodium.

References

1. Green JL, et al. *JAMA Netw Open*. 2026;9:e2559479. **2.** HHS Pain Management Best Practices Inter-Agency Task Force. Washington, DC: HHS. 2019. **3.** Qaseem A, et al. *Ann Intern Med*. 2020;173:739-748. **4.** Ailani J, et al. *Headache*. 2021;61:1021-1039. **5.** Carrasco-Labra A, et al. *JADA*. 2024;155:102-117. **6.** Dowell D, et al. *MMWR*. 2022;71:1-95. **7.** Cryer B, Feldman M. *Am J Med*. 1998;104:413-421. **8.** Warner TD, et al. *Proc Natl Acad Sci*. 1999;96:7563-7568. **9.** Young JM, et al. *Inflamm Res*. 1996;45:246-253. **10.** Blain H, et al. *Br J Clin Pharmacol*. 2002;53:255-265. **11.** Barnett J, et al. *Biochimica Biophysica Acta*. 1994;1209:130-139. **12.** Gierse JK, et al. *Biochem J*. 1999;339:607-614. **13.** Grossman CJ, et al. *Inflamm Res*. 1995;44:253-257. **14.** Kawai S, et al. *Eur J Pharmacol*. 1998;347:87-94. **15.** Kato M, et al. *JPP*. 2001;53:1679-1685.

References from the systematic search are available upon request.

Disclosures

R Mathew, S Tian, JA McGuire, D Budnitz, and A Gupta are/were employees of Kenvue Brands LLC.
S Motov and J Gudin are consultants for Kenvue but were not paid for this work.

We would like to thank Medica Communications for editorial contributions.
This study was sponsored by Kenvue Brands LLC.

Presented at PAINWeek 2026, September 8-11, 2026; Las Vegas, NV